Safety Assessment of Kavalactone-containing Herbal Drugs in Comparison to other Psychotropics

J.Schulze[1] , G.Meng[2] and C.-P.Siegers[3,4]

[1]Institute of Biometrics, UKL Luebeck; [2]Clinical Research Dr.W.Schwabe Arzneimittel,Karlsruhe; [3]Institute of Experimental and Clinical Pharmacology and Toxicology, UKL,Luebeck, Ratzeburger Allee 160, D-23538 Luebeck, Germany.

[4]Author for correspondence : siegers@medinf.mu-luebeck.de

Abstract

Currently, suspected hepatotoxicity of drug formulations contai-ning different kavolactone extracts is debated, and dependence on extraction procedures (ethanol/water, acetone/water or spissum extracts) has been postulated. More than 20 kava-containing her-bal drugs are marketed in Europe; pro duct informations mention possible adverse effects to the liver function. Even for the isolated D-L-kavaine, preclinical and clinical studies indicate hepatotoxic effects.

12 cases of suspected liver toxicity have been reported to date, for an incidence of 0.23 cases per 1 million daily doses. This number does not allow differentiation between extraction type. Additional-ly, the causality for the hepatotoxicity of kavalactones is often questionable due to comedication (other drugs, including paracet-amol), a history of alcohol abuse or viral infections. Hepatotoxic adverse effects evoked from other psychotropic drugs like benzo-diazepines, neuroleptics or antidepressants have a similar or hig-her incidence compared to kava-containing anxiolytics. Therefo-re, kavalactones do not appear to have a specific hepatotoxic profile.

Introduction

Anxiety treatment depends mainly on the use of psychotropics (benzodiazepines, doxepin) or natural products like kava extracts. Clinical improvement with kava preparations has been demon-strated in controlled studies and retrospective analysis (1). Kava pyrones (syn kava lactones) are assumed to be the active ingre-dients kava roots in extracts with acetone or ethanole. Preparations contain 70% kava pyrones (acetone/water) or 35 – 55% kava pyro-nes (ethanol/water). HPLC analysis shows that kava pyrones are similarly extracted by both procedures with identical relative amounts of the pyrones (Figure 2, HPLC trace). The main diffe-rence is the amount of inactive ingredients.

Recently, hepatotoxic reactions have been attributed to the intake of kava-containing herbal drugs (2); subsequently, some preparations have been banned from sale in Switzerland. We analyzed the incidence of hepatotoxic reactions from the BfArM database in connection with kava use, as well as the incidence of hepatic disturbances in patients summarized in the MEDIPLUS data bank.

Table 1: Mechanisms of hepatotoxicity

Some possible mechanisms of hepatic toxicity by drugs (from 3, adapted).

	Mechanism	Examples	Risk factors
Cytotoxic - Formation of reactive metabolites	exhaustion of glutathione binding of reactive metabolites to tissues	paracetamol isoniacid	enzyme induction (phenobarbitone); genetic polymorphism (N-acetyl transferase)
Metabolic interference	altered metabolic pathways of sugar and/or fatty acids	ethanol, metformin	genetic polymorphism; fatty liver degeneration
Immunologic - induction of cytotoxic antibodies	binding/adsorption of drugs to tissues Unknown	hydralazine α-methyldopa	genetic polymorphism formation of anti liver-antibodies
Cholnagitis - biliary excretion	inhibition of bile transport into canaliculi	steroids, erythromycin estolate	formation of anti liver-antibodies

Methods and Data Aquisition

We have searched the data base MEDIPLUS (IMS Health, Frank-furt) with ~1 500 000 patients. 895 847 patients had visited their physician at least once from September 1999 to August 2000; 30_000 showed signs of hepatic insult (e.g. elevated liver enzymes). Of these patients, 3075 were treated with psychoactive drugs (ATC criteria NO5A-C or NO6A, 1212 patients with sedatives and/or anxiolytics; these patients represented a total of 1313 prescriptions. The data acquisition is summarized in Figure 2. The diagnoses of liver dysfunction for the 1212 patients was fatty acid degeneration (968/1212 cases); toxic hepatosis was found in 52/1212 patients. In ~60% of the cases the time interval between drug intake and diagnosis was less than 6 months. Most prescriptions were made for the benzodiazepines bromazepam, diazepam and oxazepam, 80 cases included kava preparations (e.g. Antares, Kava-CT, Kavosporal forte, Laitan).

Drug sales for the three benzodiazepines, as well as for kava preparations, were extracted and summarized in 1000 packs; using recommended doses for anxiety, ddd sold (bromazepam 3 mg/d, diazepam 10 mg/d, oxazepam 25 mg/d, kava 120 mg/d) were calculated for the time period September 1999 to August 2000. Since for 1999 only yearly sales were available, 1/3 of the yearly sale was attributed. Total sales of these compounds adjusted to ddd were compared to incidences and estimates per 1000 ddd were calculated.

Figure 1: Data acquisition and evaluation for the incidence of hepatic dysfunction in 895 847 outpatients.

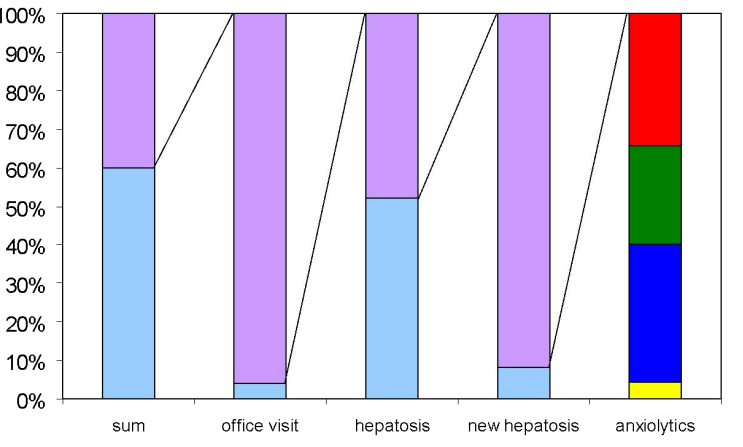
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
sum
office visit
hepatosis
new hepatosis
anxiolytics

Results

Hepatic dysfunctions and intake of anxiolytics has been correlated using the MEDIPLUS data bank containing 1 500 000 patient data. From 28 797 pati-ents with signs of hepatotoxicity, 1212 patients were treated with anxio-lytics during the last 23 months at least once (Figure 1). Most of the diagno-ses were fatty degeneration with no further specification (968/1212 cases), in 52/1212 cases toxic hepatosis was diagnosed. The incidence of hepatoto-xicity as reported to the BfArM was compared with drug sales (in 1000 pack/year). All four drugs examined showed similar incidence numbers,with bromazepam 0,06/1000 pack; diazepam 0,045/pack; oxazepam 0,038/1000 pack, kava extracts 0,044/1000 packs (compare also to incidence numbers from BfArM, Table 3).

In only about 60% of the cases, less than 6 month time elapsed between the last drug intake and signs of hepatic toxicity. This included 529 cases of fatty degeneration and 35 toxic hepatoses. No difference could be detected between the four drugs, with a similar number for kava preparati-ons (23/58 cases) than for the benzodiazepines (189/327 for bromazepam, 182/299 for oxazepam, 135/ for diazepam).

It has been suggested that the extraction procedure of kava lactones could influence the potential for adverse effects of kava preparations. The main difference between these preparations is the use of ethanol instead of acetone as the nonpolar agent (Figure 3). The resulting preparations differ in their contents of kava pyrones, with ~70% pyrones contained in acetone ex-tracts, compared to 35 – 70% pyrones in ethanolic extracts. HPLC analysis of the pyrones, as shown in the insert in Figure 3, does not indicate any dif-ference in the relative amounts of the active ingredients; for inactive com-pounds no analytical data are available. Currently there is no evidence for a major adverse potential of these constituents; thus both the pharmacological effect as well as adverse effects are attributed to the kava pyrones.

A variety of side effects has been described for kava preparations. Skin changes are well known to occur in kava drinkers in the South Pacific and Australia. A study among aborigines which had used large amounts of kava for a long time indicates major effects of kava intake (4). Among very heavy users (more than 400 g kava roots per week), elevated GT and decreased albumin levels were the only sign of hepatic toxicity, whereas urea and bilirubin levels were decreased. Among others, weight loss, distur-bances of kidney function and pulmonary hypertension was noted; however, none of these effects has been described as a result of kava pharmaceuticals. Besides elevated GT, no hepatotoxicity has not been found in this cohort. Albeit genetic differences can be expected between caucasians and australian aborigines, and cytochrome P4502D6 deficiency has been found in patients with kava associated liver toxicity (2); these data confirm the low toxic potency.

Table 2: Incidence of adverse effects reported for anxiolytics

Reported adverse effects were extracted from the drug information and graded according to severe [++], occasional [+], rare [(+)] and not reported [0]. Drug interactions: dynamic – pharmacodynamic interactions; kinetic: interference with drug metabolism

Adverse effect	Kava extract Oxazepam		Bromazepam Doxepin	Diazepam	
Gastrointestinal	(+)	+	+	+	++
CNS	(+)	+	+	+	++
Hepatotoxicity	(+)	(+)	(+)	(+)	++
Drug interactions	dyn.	Dyn./kin.	Dyn./kin.	Dyn./kin.	Dyn.
Circulation	0	(+)	(+)	(+)	++
Muscle	0	(+)	(+)	(+)	++
Allergy	(+)	(+)	(+)	(+)	(+)
Addiction	0	+	+	+	(+)
Reproduction	unknown unknown				

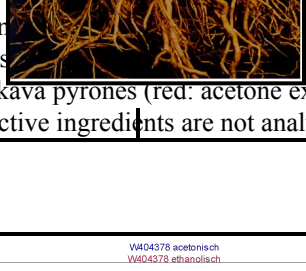

Figure 2: Kava preparatio[illegible]n

The figure shows the main [illegible]thanolic and acetonic kava extracts [illegible] presence and amounts of kava pyrones (red: acetone extract; blur: ethanol extract); inactive ingredients are not analyzed.

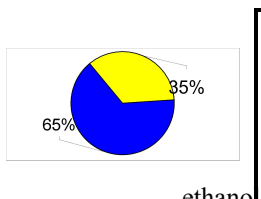
35%
65%

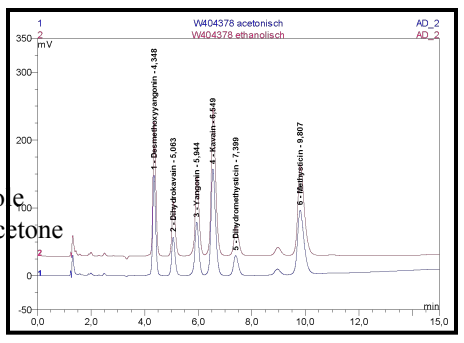
1 W404378 acetonisch AD_2
2 W404378 ethanolisch AD_2
mV
350
300
200
100
0
-50
1 - Desmethoxyyangonin - 4,348
2 - Dihydrokavain - 5,063
3 - Yangonin - 5,944
4 - Kavain - 6,549
5 - Dihydromethysticin - 7,399
6 - Methysticin - 9,807
0,0
2,0
4,0
6,0
8,0
10,0
12,0
15,0
min

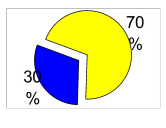
70 %
30 %

ethanole

acetone

Table 3: Incidence of hepatotoxicity for anxiolytic compounds

The table lists hepatotoxicity incidences including toxic hepatosis in MEDIPLUS data base patients in association with anxiolytic intake (per 1000 packssold, or per 1 million ddd).

	Inzidenz (n/1000 packs)	Inzidenz (n/1000000ddd)
Bromazepam	0.062	0.90
Diazepam	0.054	2.12
Oxazepam	0.038	1.23
Kava (ethanol and acetone extracts)	0.044	0.89

Table 3 shows relative incidence numbers of hepatotoxicity associated with the intake of anxiolytic drugs. The data indicate a comparable, low potential of hepatic adverse effects in correlation with the intake of both benzodiazepines and kava preparations. Oxazepam has an incidence for hepatotoxic adverse effects of 1.2, diazepam 2.1, kava preparations (both ethanolic and acetonic extracts) and bromazepam 0.9 cases/million ddd during the time period 9/1999 to 8/2000; these incidence do not appear to be substantially different given the uncertainty of association between adverse effect an drug intake. If incidence numbers are calculated based on 1000 packs sold, again there is no difference between different anxiolytics (Table 3).

It should be noted that some kava preparations are OTC drugs and thus may not be present in the sales figures obtainable; also benzodiazepine adverse effects are well known; thus an underreporting may occur. On the other hand, most cases of hepatosis as seen in the Mediplus base bank likely are caused by other factors, e.g. alcohol or fat intake.

Some drugs registered for use as anxiolytics by the BfArM, as well as their major properties are listed in Table 4; a comparison indicates no “safe” anxiolytic presently available.

Table 4: Therapeutic alternatives in anxiolytic treatment

--

-Benzodiazepines long acting drugs ($t_{½}$ 6 – 24h); not indicated in hyopotonia, paradoxic drug reaction, in-teraction with e.g. coumarin

Kava preparations short acting substances, not indicated in children, no experience in pregnancy and lactation. Rarely drug interactions with cen-trally acting compounds.

Hypericum short acting substances, not indicated in children, no experience in pregnancy and lactation. Interaction with e.g. coumarins

Doxepin antidepressant, common and serious adverse effects, not indicated as first line therapy.

--

Conclusions

- Hepatotoxicity is a rare adverse effect for kava prepara-tions, as well as for benzodiazepines used as anxiolytic compounds. Other drugs listed as anxiolytics have compa-rable (benzodiazepines) or more severe (doxepine) adverse effects.
- Data from both spontaneous reporting (BfArM) or an out-patient data base (MEDIPLUS) do not indicate differences in the incidence of hepatotoxicity, when stratified to ddd sold.
- Reports from the BfArM data base indicate association of hepatotoxicity with both acetone and ethanole extracts, with no obvious differences between these extractions.
- Kava pyrones are available as OTC preparations. Thus the true intake may be underestimated as compared to prescrip-tion drugs like benzodiazepines. Benzodiazepines have a well known potential for adverse effects (paradoxic effects, sedation), which also may not be reported spontaneously.
- Currently there is no evidence for higher toxicity of either ethanolic or acetonic kava preparations.

References

1. Pittler MH; Ernst E (2000) Efficacy of kava extract for treating anxiety: systematic review and meta-analysis. J. Clin. Psychopharmacol. **20**, p. 84-82

2. Russmann S, Escher M, Stoller R, Lauterburg BH (2001) Hepatotoxicity of kava (piper mystheticum) – containing herbal drugs – recent cases in Switzerland and investigations regarding the mechanism. NS Arch. Pharmacol. ##

3. Isselbacher, et al: Harrison’s Principles of Internal Medicine. 13th edition. McGraw Hill, 1997

4. Matthews, J.D. et al. (1998) Effects of the heavy usage of kava on ohysical health: summary of a pilot survey in an Aboriginal community. Med. J. Austr. 148, S. 548 - 555